Functional Rescue of CFTR by Aerosolized Delivery of Optimized CFTR mRNA Using ReCode LNPs in Primary Human Bronchial Epithelial Cells Derived From Patients With Cystic Fibrosis

Daniella Ishimaru[1], Dmitri Boudko[1], Ella A. Meleshkevitch[1], Maninder S. Sidhu[1], Julia R. Poniatowski[1], Peiyang Gao[1], Touhidul I. Molla[1], Sierra R. Comini[1], Harriet E. Lister[1], Melissa L. Coquelin[1], Crystal Johnson[1], Ali Alfaifi[1], Omid M. Mousa[1], Xueliang Yu[1], Rumpa B. Bhattacharjee[1], David Liston[1], Jackson K. Eby[1], Mirko Hennig[1], Robert J. Bridges[2], Philip J. Thomas[3], Vladimir G. Kharitonov[1], Brandon A. Wustman[1], David J. Lockhart[1], Michael J. Torres[1]

[1]ReCode Therapeutics, Inc, Dallas TX and Menlo Park, CA, United States; [2]Rosalind Franklin University of Medicine and Science, North Chicago, IL, United States; [3]University of Texas Southwestern Medical Center, Dallas, TX, United States

RATIONALE

Cystic fibrosis (CF) is a progressive, genetic disease affecting a chloride channel, CFTR, located on the apical plasma membrane of specialized epithelial cells. Defective pulmonary mucociliary clearance (MCC) is one of the main phenotypes in CF patients. Because a significant fraction of the CF patient population is not amenable to currently approved CFTR modulators such as Trikafta, the search for universally applicable therapies that promote CFTR function and mucus clearance remains a goal. To address this challenge, ReCode Therapeutics is advancing an mRNA-based treatment to rescue CFTR function using its proprietary lipid nanoparticle (LNP) platform and optimized CFTR mRNA sequences delivered as an inhaled aerosol.

ReCode's sequence optimization of CFTR mRNA eliminates hydrolysis hot-spot

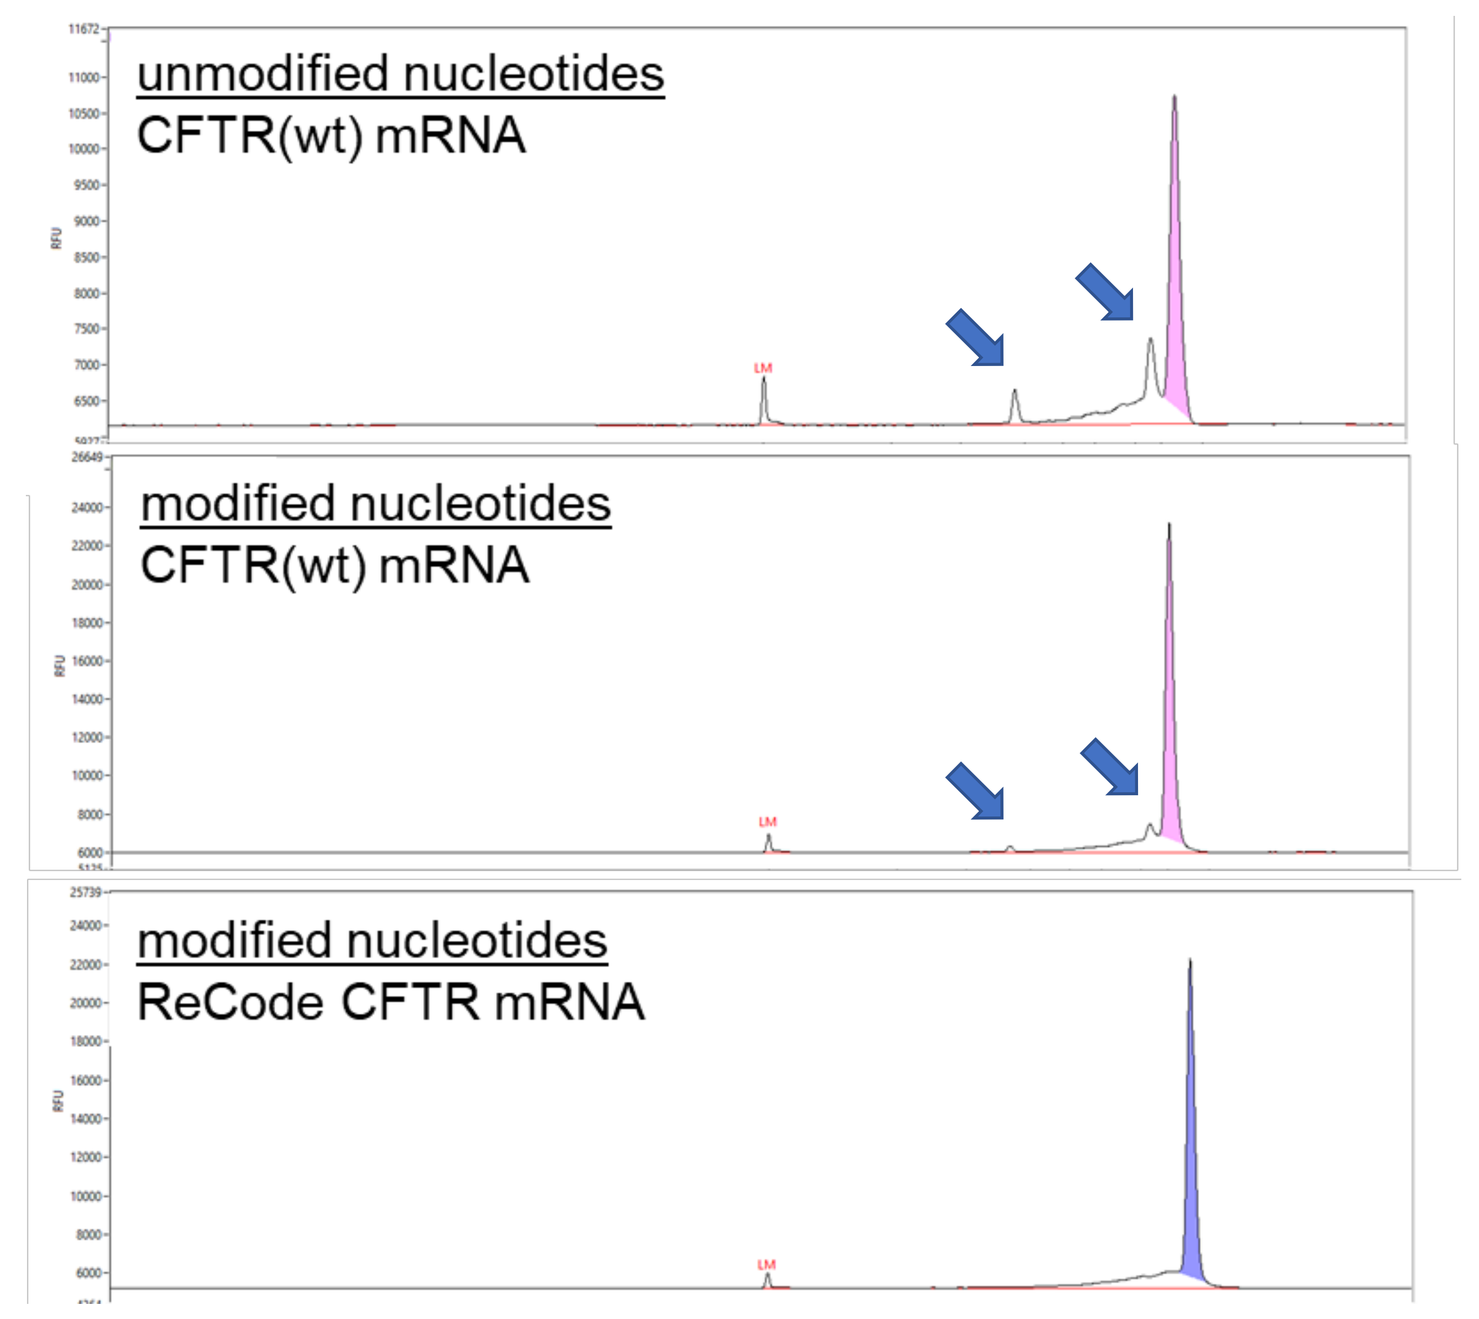
unmodified nucleotides
CFTR(wt) mRNA
modified nucleotides
CFTR(wt) mRNA
modified nucleotides
ReCode CFTR mRNA

Native or ReCode-optimized CFTR mRNAs were *in vitro* transcribed in the presence of unmodified or modified nucleotides, where indicated. Purified samples were analyzed on a Fragment Analyzer (Agilent). Larger peak corresponds to full-length CFTR mRNA. A specific hydrolysis hot-spot (arrows) was observed with the wild-type, native CFTR mRNA sequence. *In vitro* transcription of CFTR(wt) mRNA in the presence of modified nucleotides reduced the observed hydrolysis (middle panel). Combination of ReCode's optimized sequence and modified nucleotides allowed for synthesis of full-length CFTR mRNA (bottom panel).

Evaluation of modified nucleotides and selection of a lead mRNA

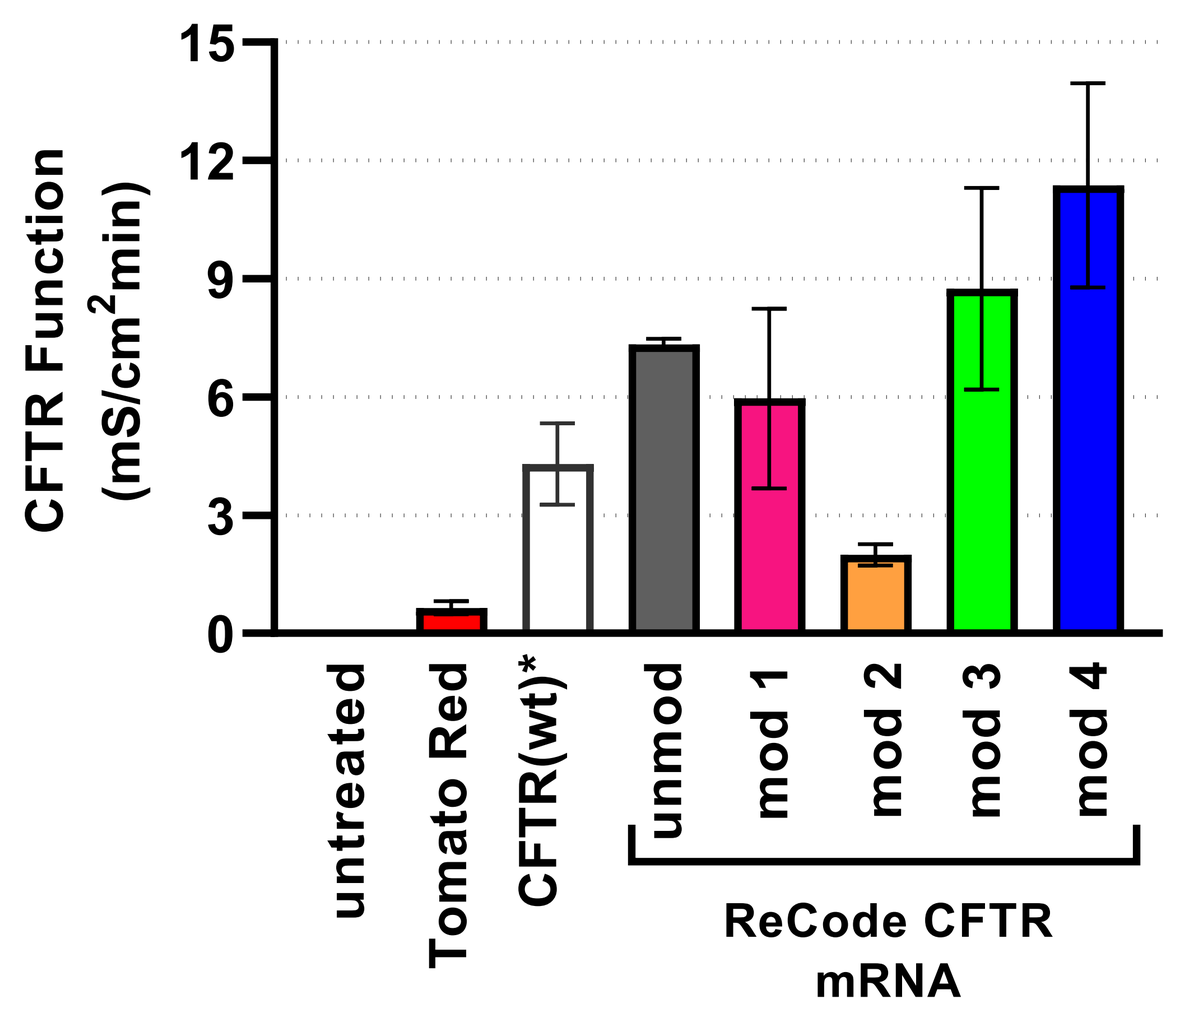
CFTR Function (mS/cm²min)
untreated
Tomato Red
CFTR(wt)*
unmod
mod 1
mod 2
mod 3
mod 4
ReCode CFTR mRNA

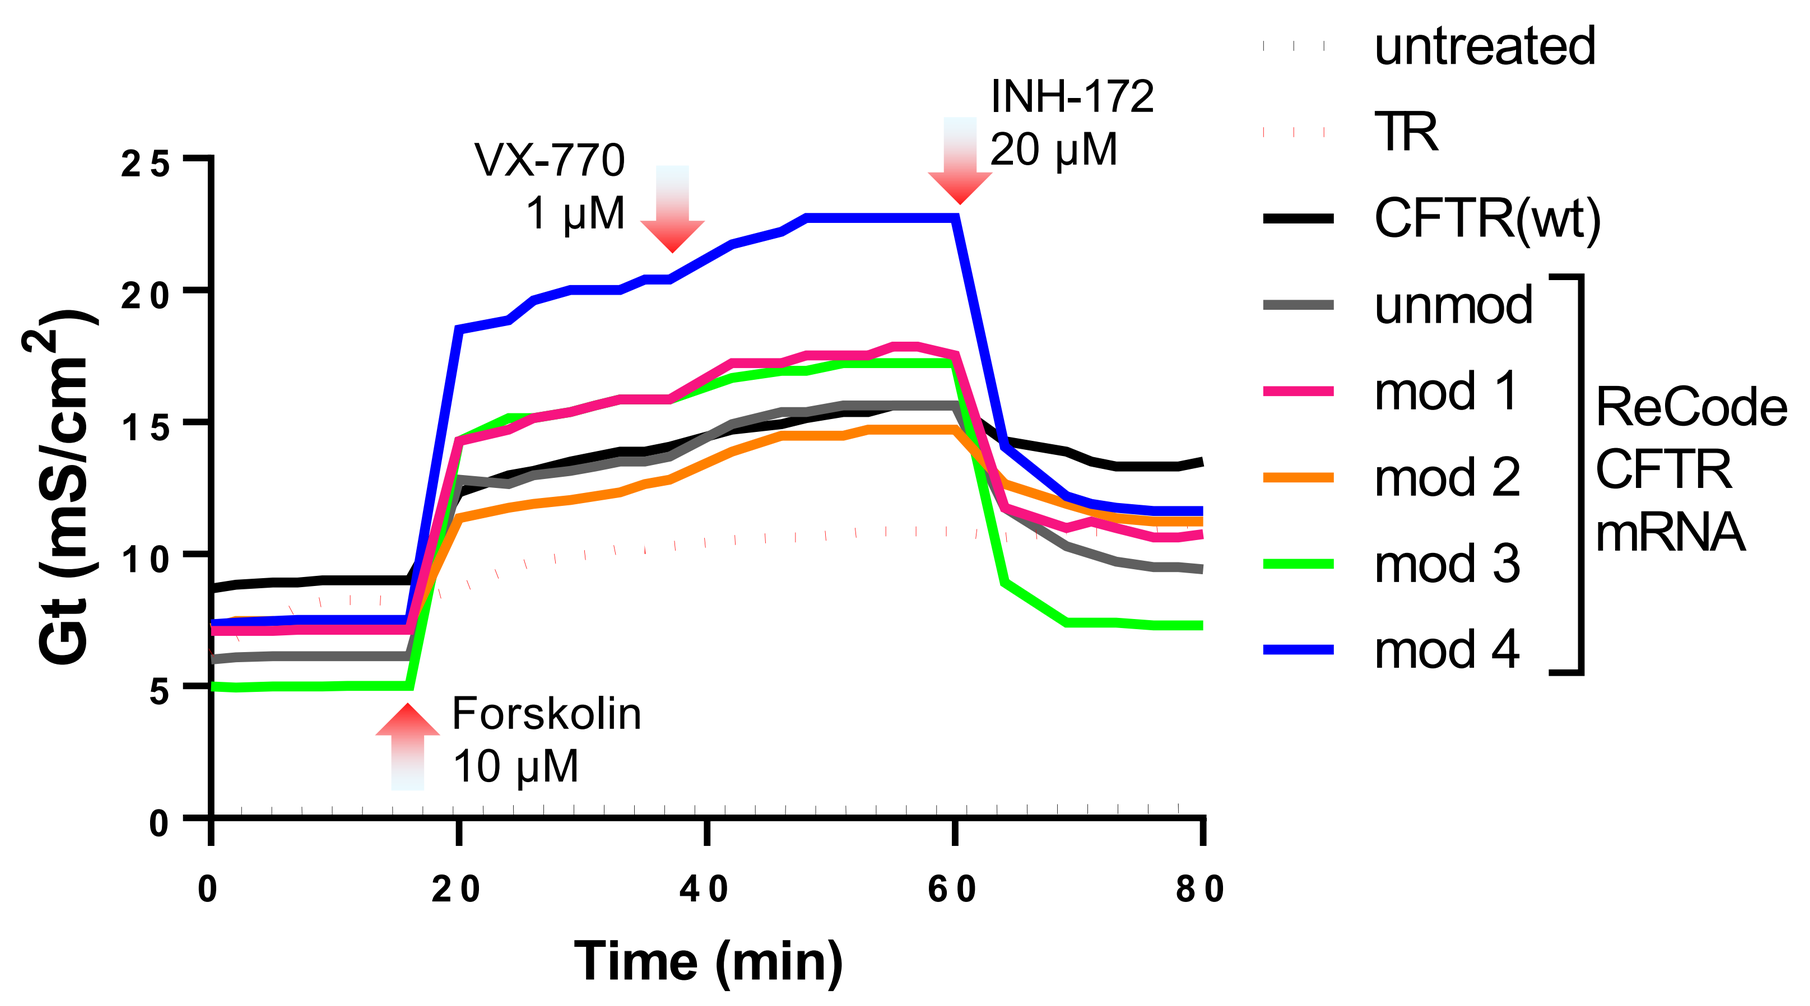
Gt (mS/cm²)
Time (min)
VX-770 1 µM
INH-172 20 µM
Forskolin 10 µM
untreated
TR
CFTR(wt)
unmod
mod 1
mod 2
mod 3
mod 4
ReCode CFTR mRNA

Three days old confluent FRT cells grown on TransWell permeable support were transfected with different CFTR mRNAs using Lipofectamine 2000. MTECC24 assay of the transepithelial conductance was performed 24h post-transfection. On the left, transepithelial conductance (Gt) responses, bars are Gt-area under the curve per min between Forskolin and INHibitor-172 addition time-points. On the right, representative conductance kinetic traces in FRT monolayer after addition of CFTR modulators, as indicated. We identified a CFTR mRNA sequence and composition with improved stability that is more efficiently translated into functional CFTR protein compared to the wild-type sequence, both in FRT and primary hBE cells (not shown). *Note: the CFTR(wt) mRNA was transcribed in the presence of modified nucleotides to favor the presence of full-length mRNA. Utilization of modified nucleotides also lead to reduced immunoreactivity *in vitro* (see poster P316: Optimization of DNAI1 mRNA Constructs to Treat Primary Ciliary Dyskinesia).

ReCode's proprietary formulation A (LNP A) is the top performer in G542X/ΔF508 hBE cells

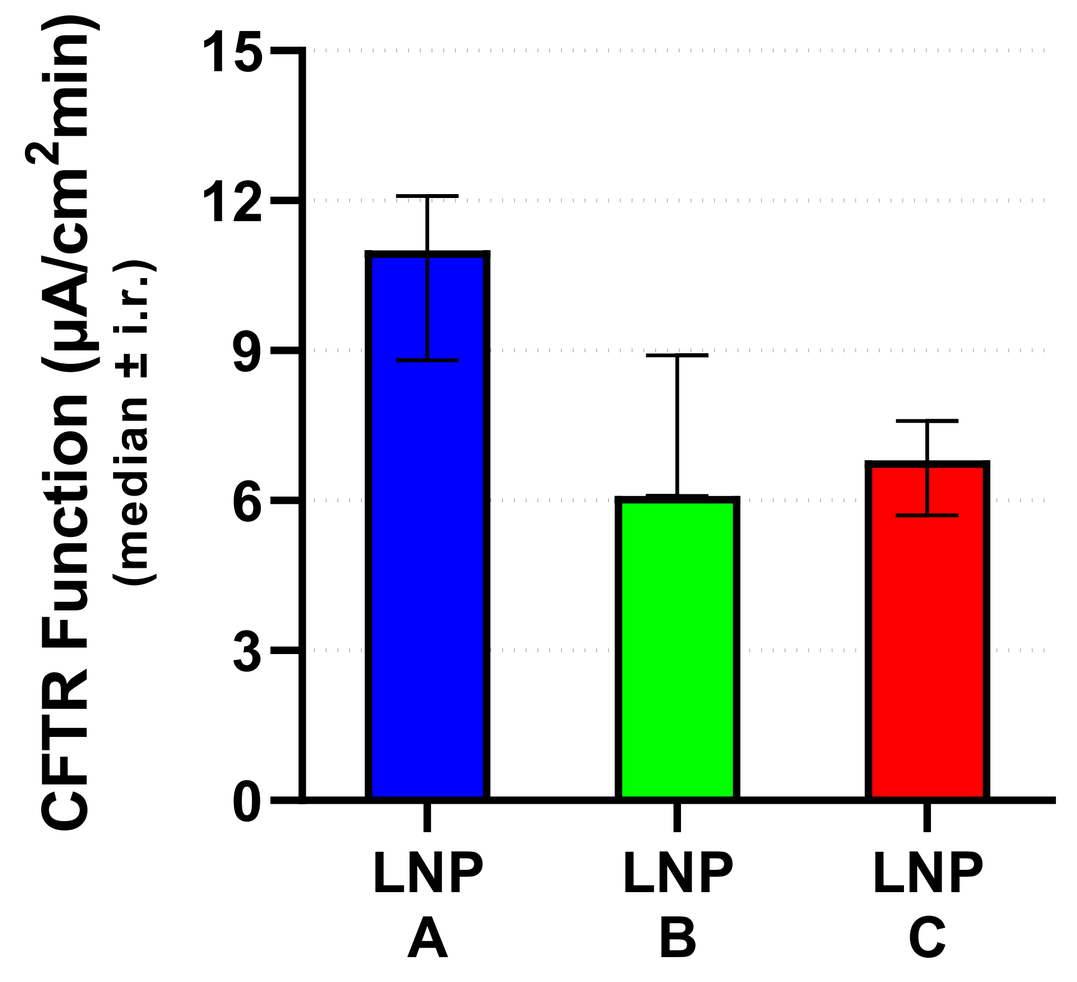
CFTR Function (µA/cm²min)
(median ± i.r.)
LNP A
LNP B
LNP C

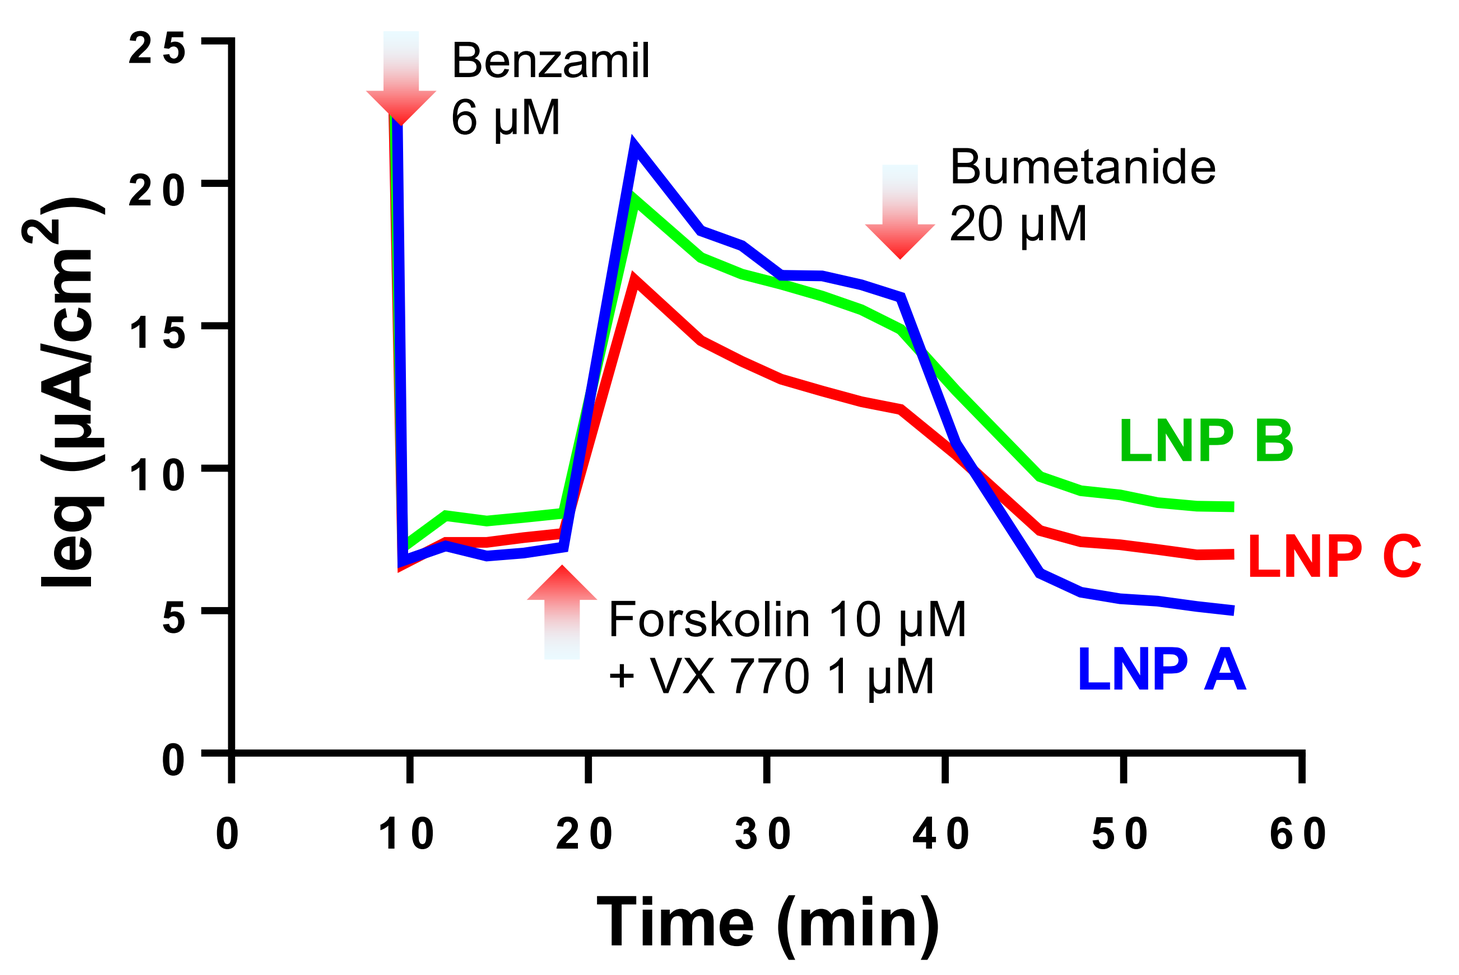
Ieq (µA/cm²)
Time (min)
Benzamil 6 µM
Bumetanide 20 µM
Forskolin 10 µM + VX 770 1 µM
LNP B
LNP C
LNP A

G542X/ΔF508 CF patient-derived, human bronchial epithelial (hBE) cells were grown at an air-liquid interface. Fully differentiated cells were aerosolized using a commercially available mesh nebulizer with ReCode's LNPs containing CFTR mRNAs. On the left panel, CFTR function was determined as forskolin-induced and bumetanide-suppressed current measured with robotic transepithelial current clamp system (MTECC24). Representative traces are shown on the right. ReCode's LNP A-formulated CFTR mRNA shows the highest rescue of CFTR function.

Higher doses of LNP A formulated CFTR mRNA are well-tolerated in G542X/ΔF508 cells

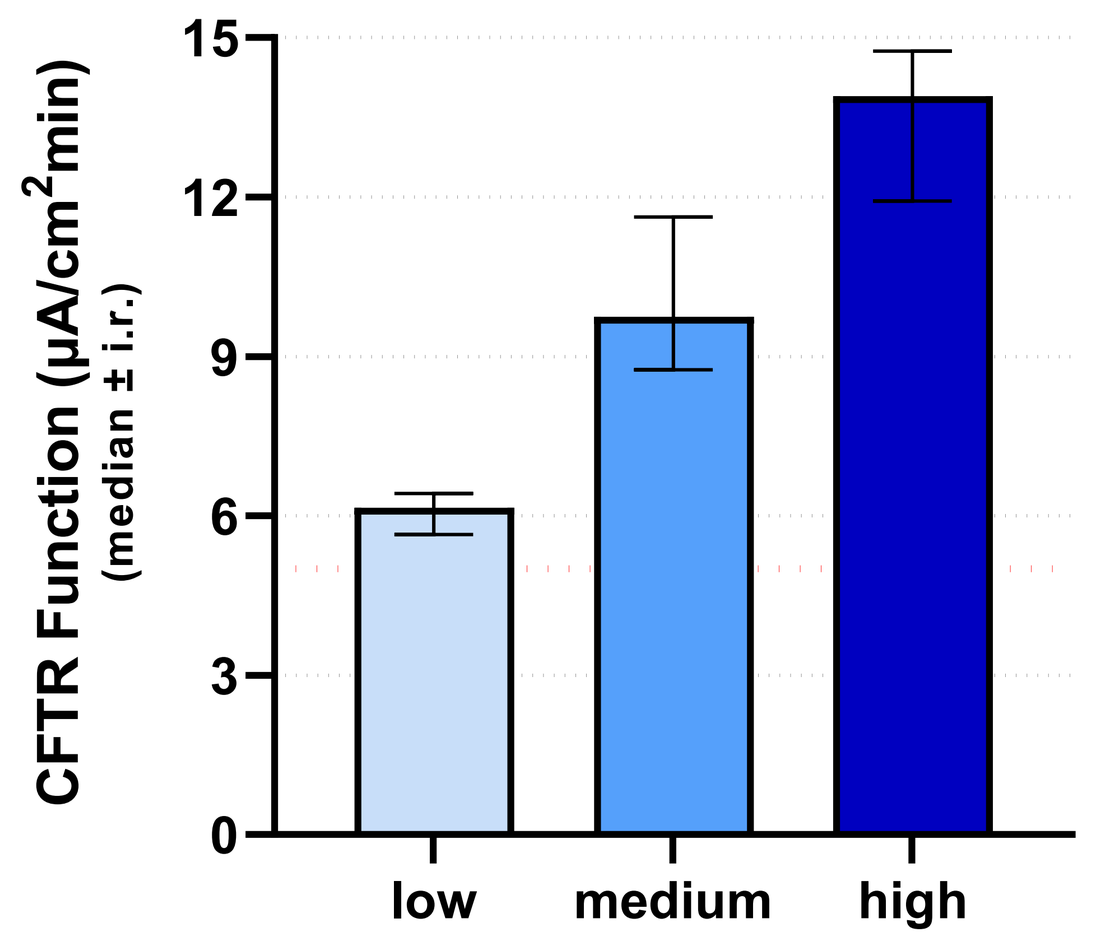
CFTR Function (µA/cm²min)
(median ± i.r.)
low
medium
high

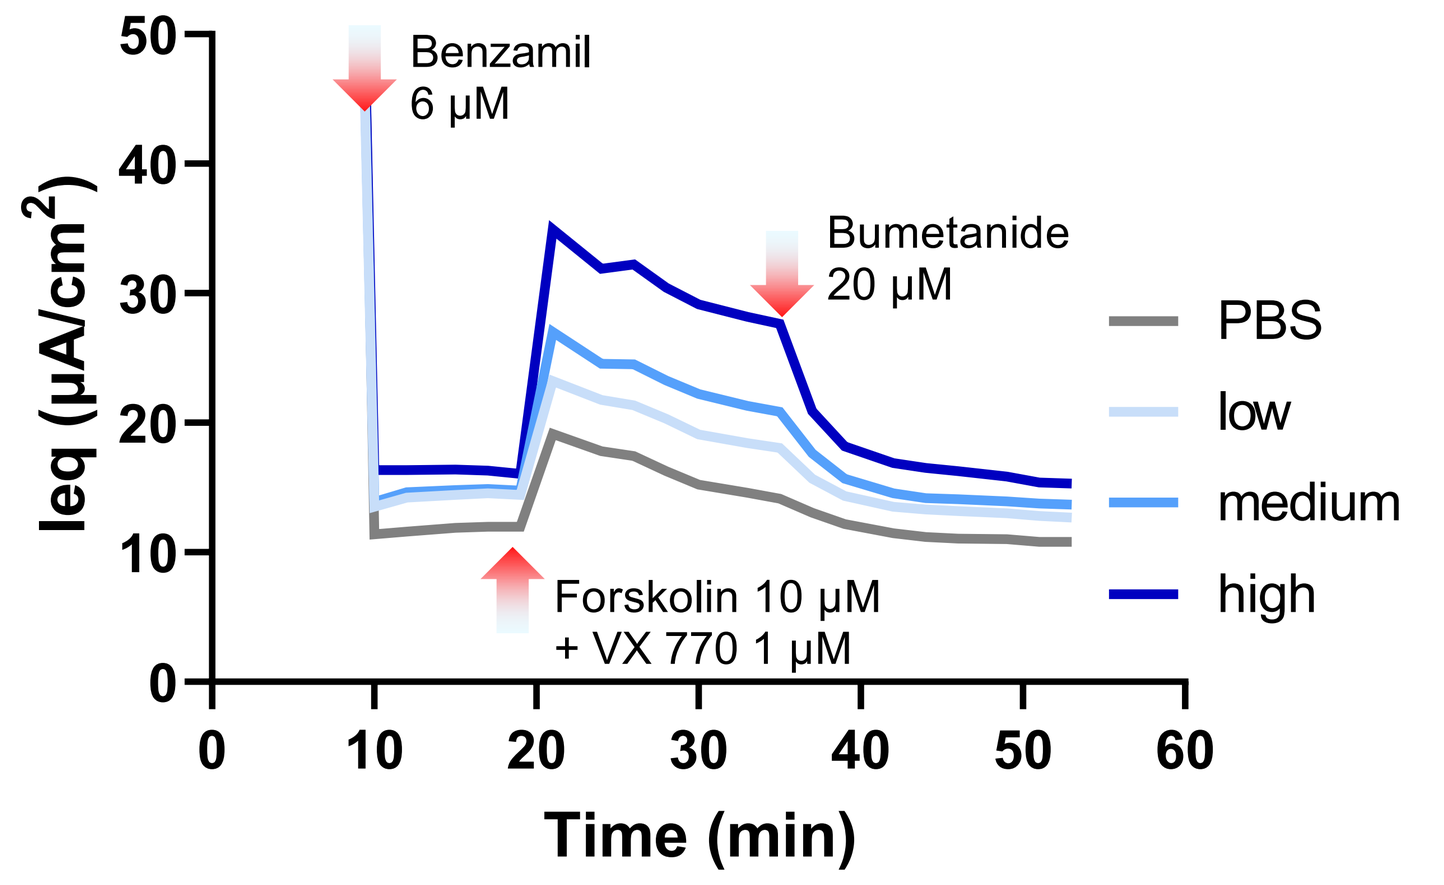
Ieq (µA/cm²)
Time (min)
Benzamil 6 µM
Bumetanide 20 µM
Forskolin 10 µM + VX 770 1 µM
PBS
low
medium
high

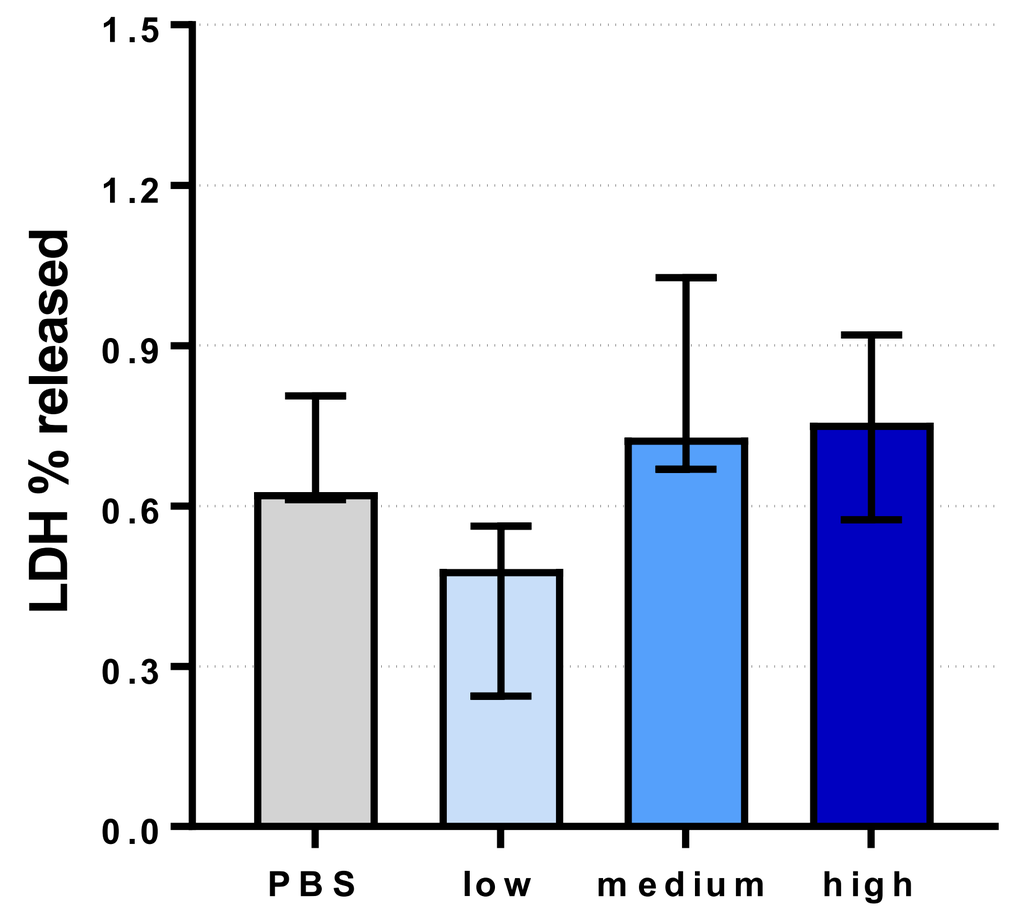
LDH % released
PBS
low
medium
high

Fully differentiated cells were nebulized with low, medium, or high doses of HA-CFTR mRNA-formulated with ReCode's proprietary formulation A (LNP A). Analysis was performed 24h post-nebulization. CFTR function was determined as described above. On the top right panel, representative traces. Bottom left, Percentage of LDH release relative to whole cell lysate (= 100%). For reference, historical data with cytotoxic LNPs showed values for released LDH at ~60%. Here we show that even at the highest dose delivered, no cytotoxicity was detected with ReCode's LNP. TEER measurements show similar values among all conditions (>300 Ω/cm²).

CFTR mRNA restores function in CF patient-derived hBE cells including genotypes not responsive to modulators

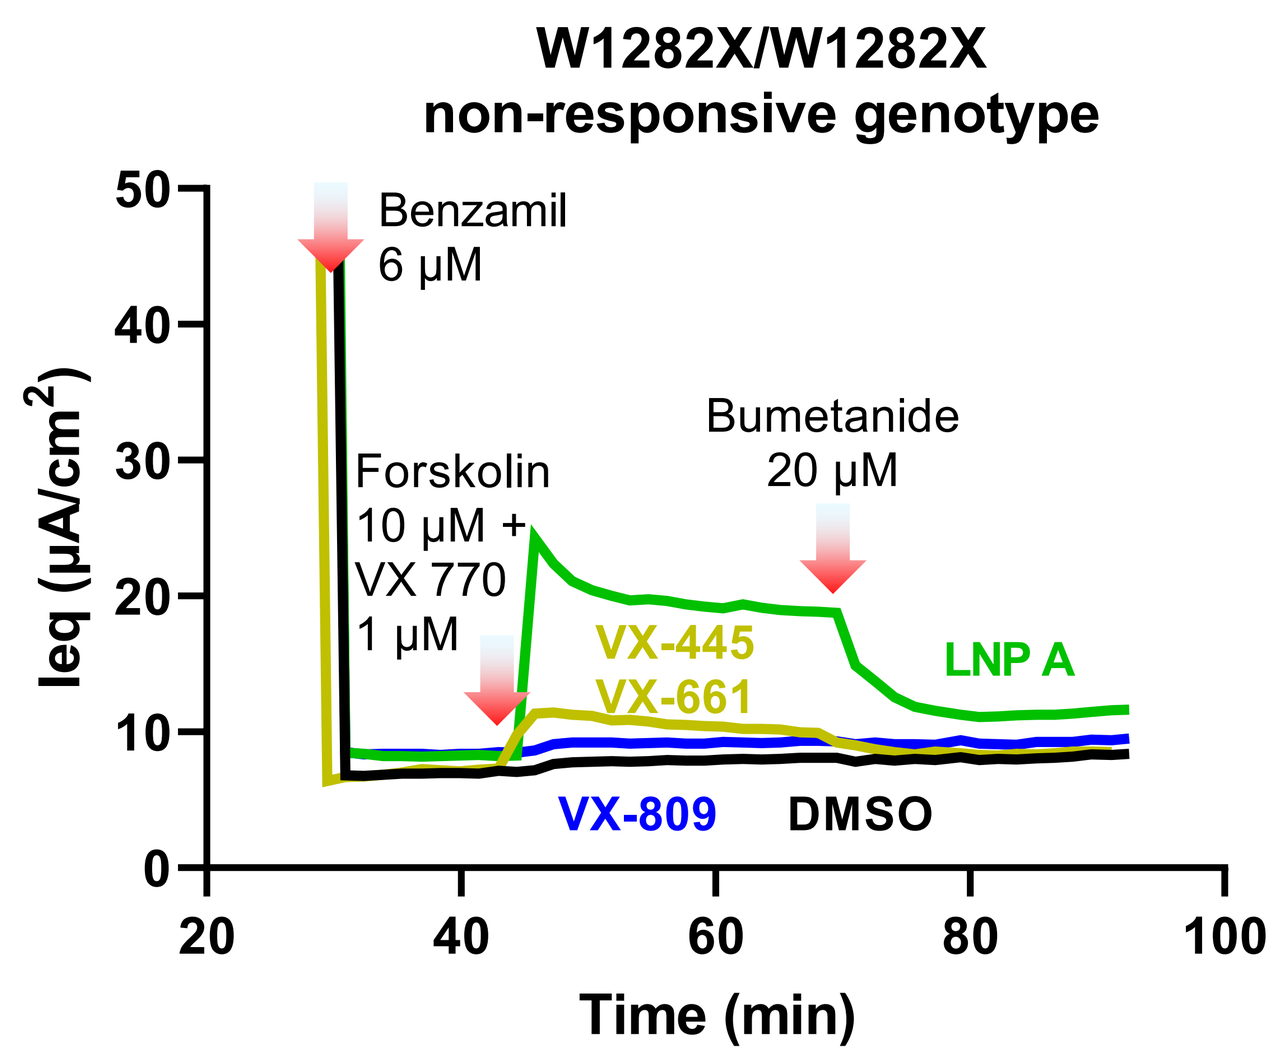
W1282X/W1282X
non-responsive genotype
Ieq (µA/cm²)
Time (min)
Benzamil 6 µM
Forskolin 10 µM + VX 770 1 µM
Bumetanide 20 µM
VX-445
VX-661
LNP A
VX-809
DMSO

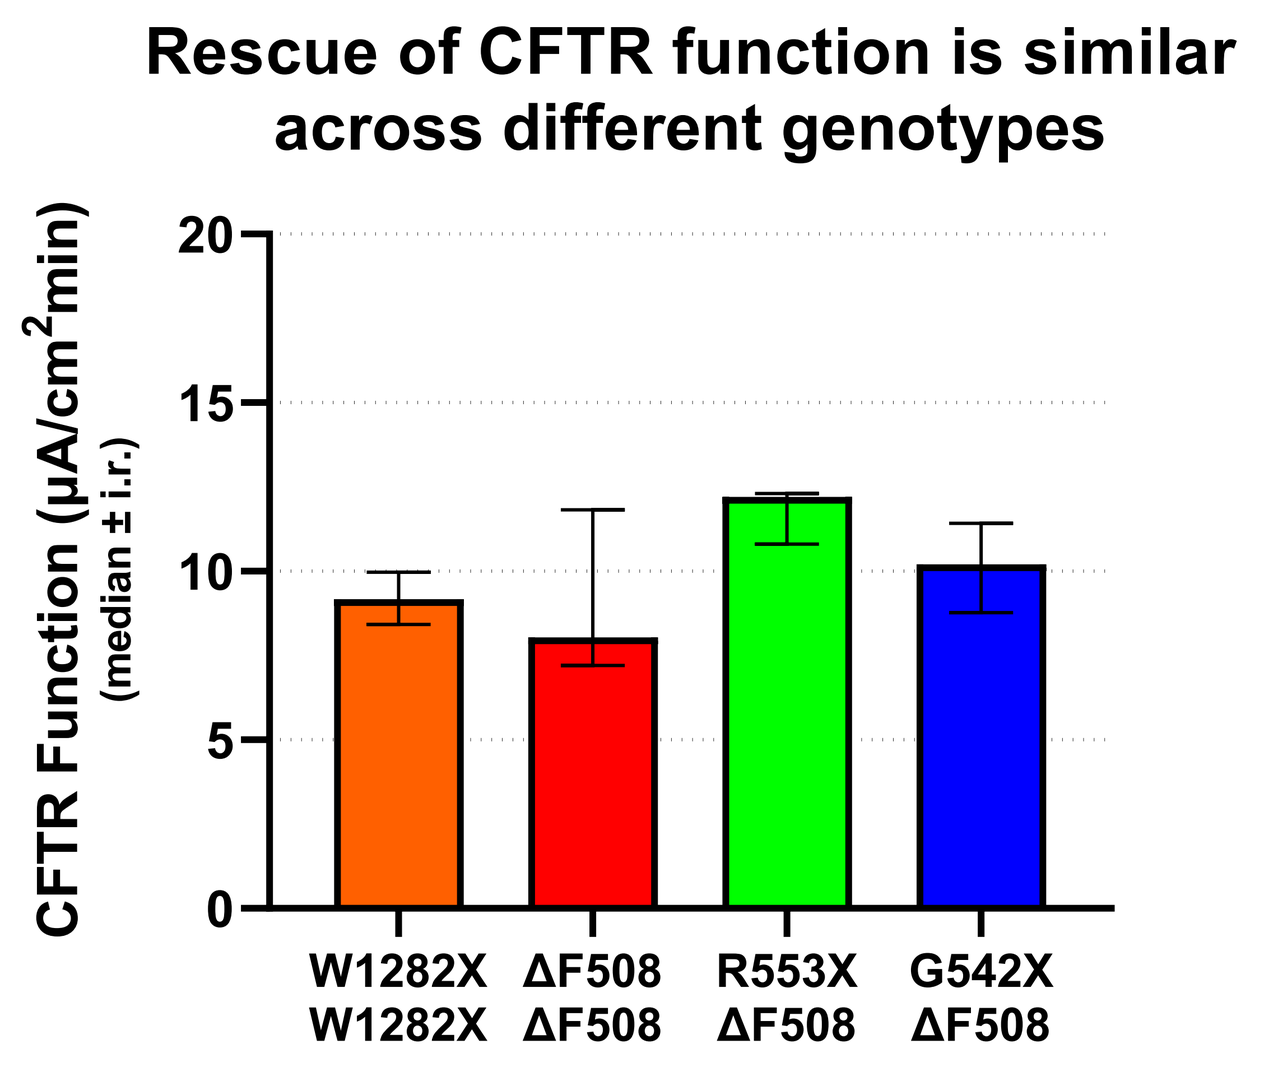
Rescue of CFTR function is similar across different genotypes
CFTR Function (µA/cm²min)
(median ± i.r.)
W1282X W1282X
ΔF508 ΔF508
R553X ΔF508
G542X ΔF508

hBE cells with Trifakta-responsive and non-reponsive CF genotypes were grown on permeable support in 24 well TransWell plate. Fully differentiated cells were nebulized with high dose of HA-CFTR mRNA formulated with ReCode's proprietary formulation A (LNP A). Analysis was performed 24h post-nebulization. CFTR function was determined as described before. On the left panel, representative traces for the W1282X/W1282X genotype.

Transfection of hBEs with Td tomato mRNA reveals formulation-specific cell tropism signatures

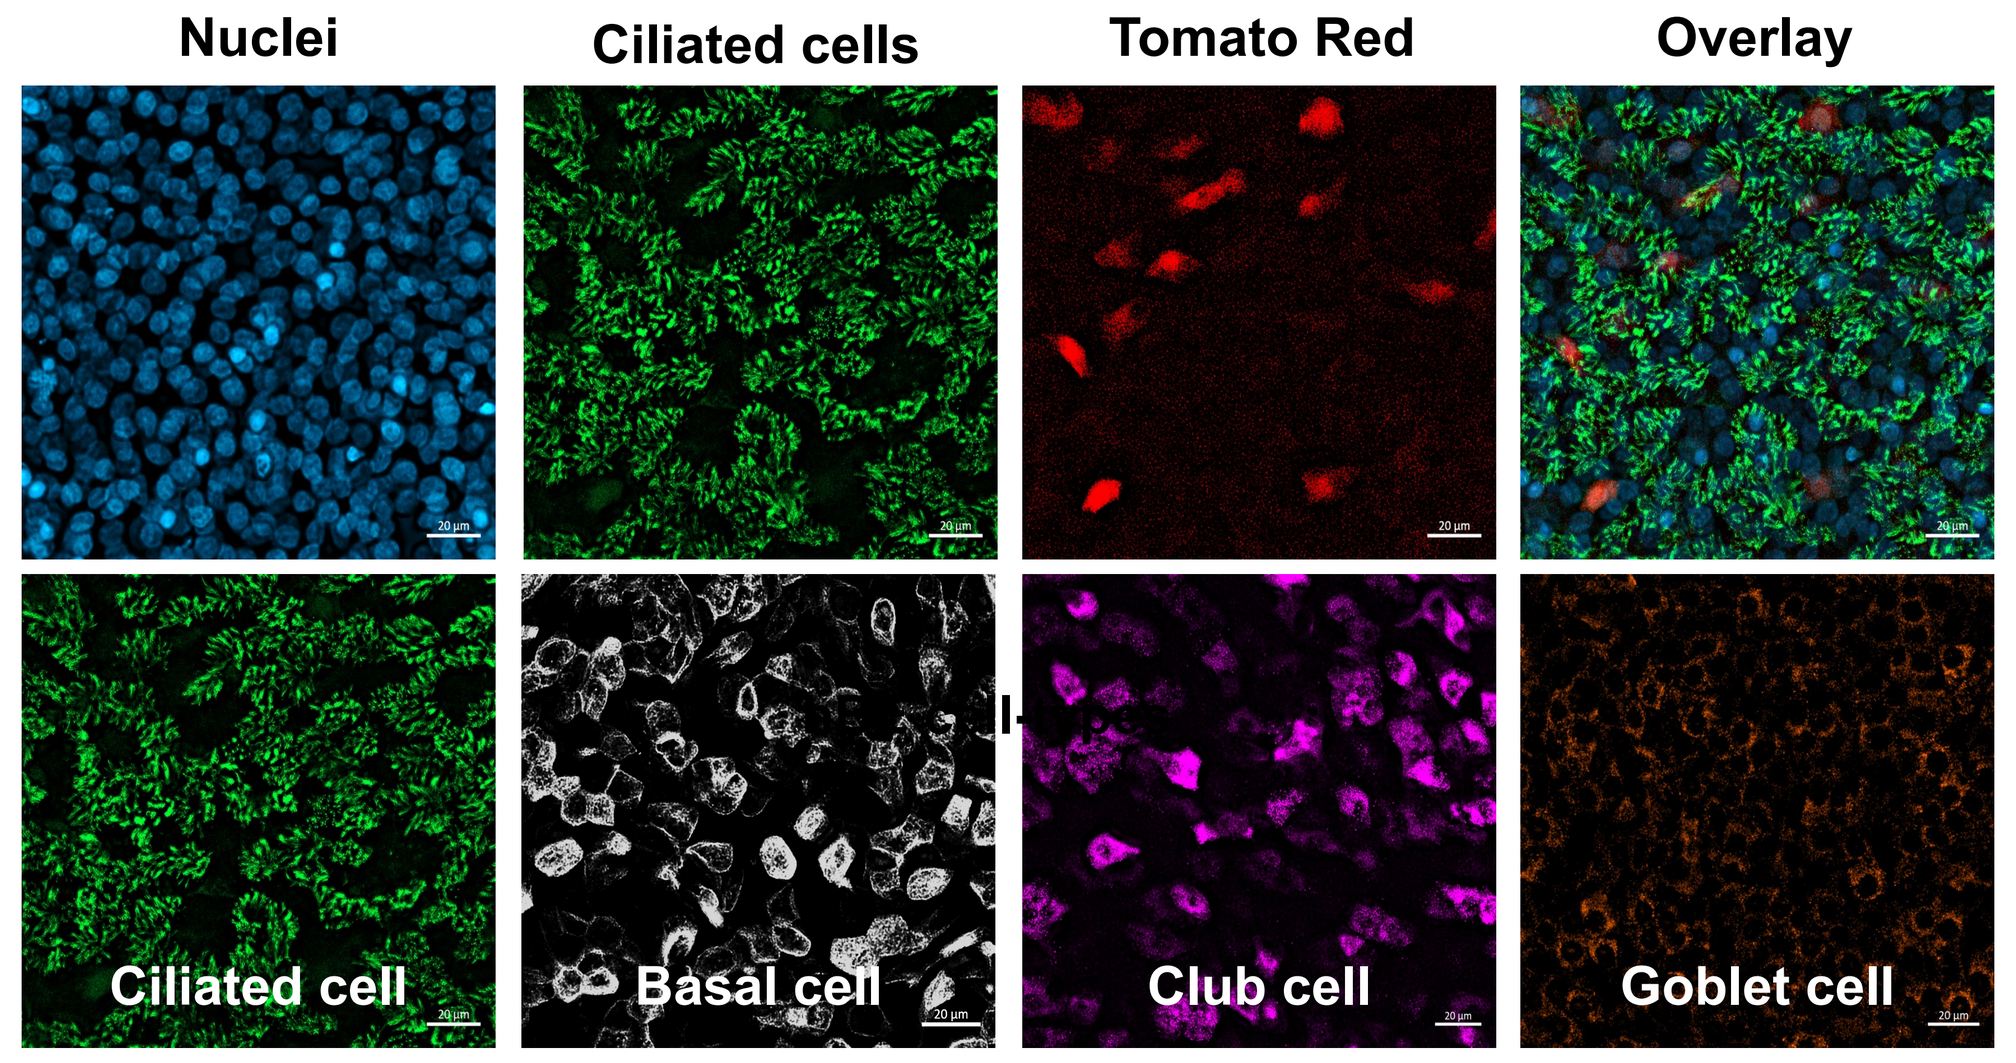
Nuclei
Ciliated cells
Tomato Red
Overlay
Ciliated cell
Basal cell
Club cell
Goblet cell

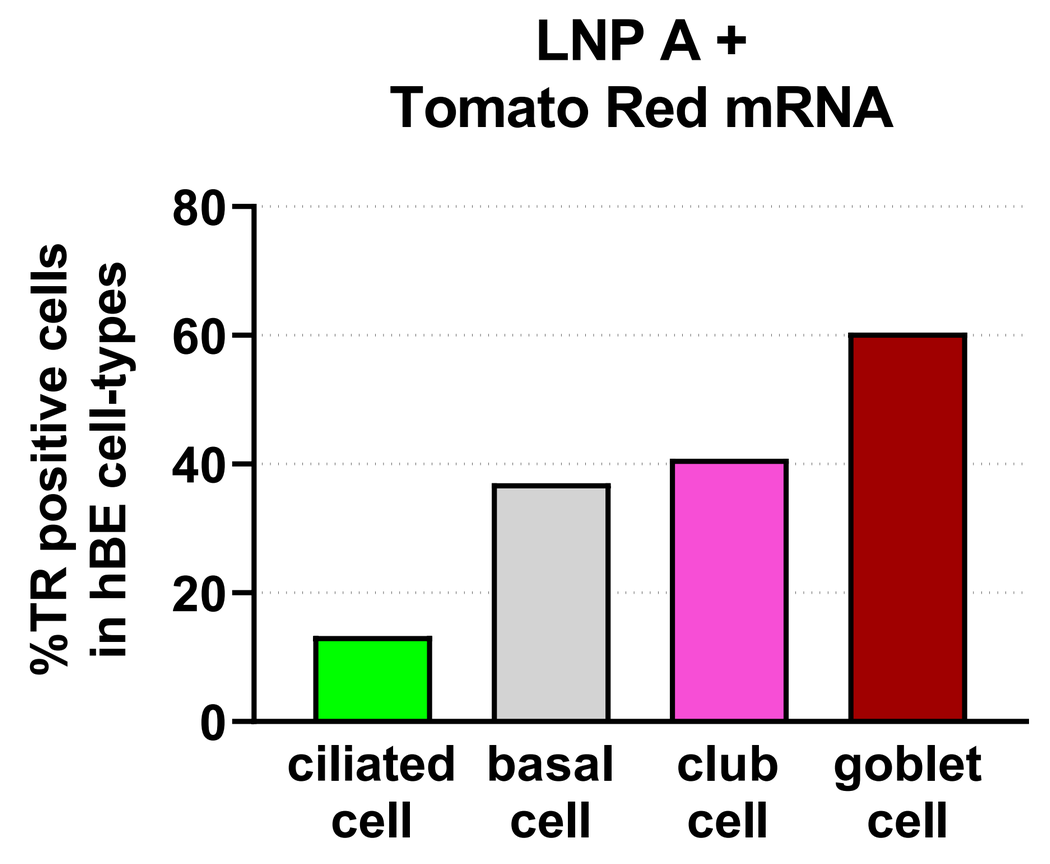
LNP A +
Tomato Red mRNA
%TR positive cells in hBE cell-types
ciliated cell
basal cell
club cell
goblet cell

Cells were fixed, blocked, and probed with primary antibodies for each cell type (for ciliated cells: anti-acetylated tubulin - TUBA; for basal cells: anti-cytokeratin 5 - CK5; for club cells: anti-secretoglobin family 1A member 1 - SCGB1A1 and anti-CC10; for goblet cells: anti-mucin 5AC – MUC5AC). Images were taken using Zeiss Microscope, Axio observer 7 at 40x (7 fields) in two channels (555 and 647 nm); Z stack images were taken. Compressed images were taken only for ciliated cells to accommodate cells in different planes. For every field, total number of cells with TR signal and colocalized signal are counted. Percent of positive Tomato Red signal in each cell type was determined.

Bar graph: Fully differentiated wild-type hBE cells were treated once with ReCode's proprietary formulation A (LNP A)-formulated Td Tomato mRNA (4 µg) using Vitrocell nebulization. Percentage of positive cells were determined by co-localization with each cell type marker as shown in the images.

CONCLUSIONS

Our results demonstrate the capability of the ReCode's LNP platform to deliver optimized, functional CFTR mRNA in well-differentiated CF hBE cultures as an aerosol. These preclinical data support further investigation and provide a practical approach to address a significant patient population that does not benefit from current CFTR modulator therapy.

DISCLOSURES

(Ishimaru) Research supported by – ReCode Therapeutics, Authors relevant interests – ReCode Therapeutics, Employee and hold stock options

(Boudko, Meleshkevitch, Sidhu, Poniatowski, Gao, Molla, Comini, Lister, Coquelin, Johnson, Alfaifi, Mousa, Yu, Bhattacharjee, Liston, Eby, Hennig, Kharitonov, Wustman, Lockhart, Torres) Research supported by – ReCode Therapeutics, Authors relevant interests – ReCode Therapeutics, Employee and hold stock options

(Bridges, Thomas) Research supported by – ReCode Therapeutics, Authors relevant interests – ReCode Therapeutics, Consultant and hold stock options

Part of this work was sponsored by grant number RECODE17X0-SC from the Cystic Fibrosis Foundation.

NO PICTURES OR RECORDING ARE ALLOWED